#615

Targeted Immune Cell Activation by Systemic Delivery of Toll-Like Receptor 9 Agonist Antibody Conjugates Induce Potent Anti-Tumor Immunity

Ons Harrabi[1], Amy Chen[1], Emma Sangalang[1], Laura Doyle[1], Danielle Fontaine[1], Bora Han[1], Curt Bradshaw[2], Jaume Pons[3], Janet Sim[1], Tracy C. Kuo[1], Hong I. Wan[1]

[1]Tallac Therapeutics, Inc., Burlingame, CA; [2]Arrowhead Pharmaceuticals, Pasadena, CA; [3]ALX Oncology, Burlingame, CA

Introduction

- Despite the success of checkpoint inhibitors and adoptive CAR-T therapies, only a fraction of cancer patients benefit. Novel therapies engaging both innate and adaptive immune response may produce more robust and durable anti-cancer immunity [1].
- Toll-like receptors (TLRs) play crucial roles in mounting potent innate immune responses against invading pathogens. Activation of TLR signaling leads to induction of inflammatory cytokines and priming of adaptive immunity [2].
- TLR9 agonists have shown clinical responses in melanoma patients by intra-tumoral injection [3].
- We developed a novel Toll-like Receptor Agonist Antibody Conjugate (TRAAC) platform to deliver a potent TLR9 agonist (T-CpG) for targeted immune activation via systemic administration.
- We evaluated various TRAAC approaches targeting either immune cell receptors or tumor specific antigens, demonstrating robust immune modulation and potent single agent anti-tumor activity in pre-clinical settings.

Fig 1: TRAAC is a Toll-Like Receptor Agonist Antibody Conjugate platform intended for systemic delivery of T-CpG, a potent TLR9 agonist

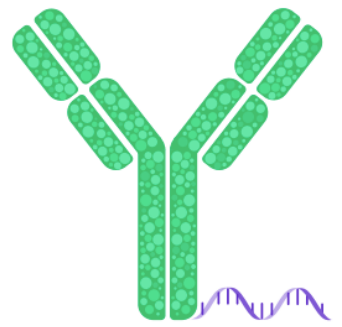

Antibody: Versatile TRAAC targeting
- **Specific immune cells, or**
- **Tumor microenvironment**

Immune Activator: T-CpG
- **Potent TLR9 agonist**
- **Linear, monomeric, and non-aggregated**
- **Sequence optimized for potency and stability**
- **Innovative design enables efficient site-specific conjugation**

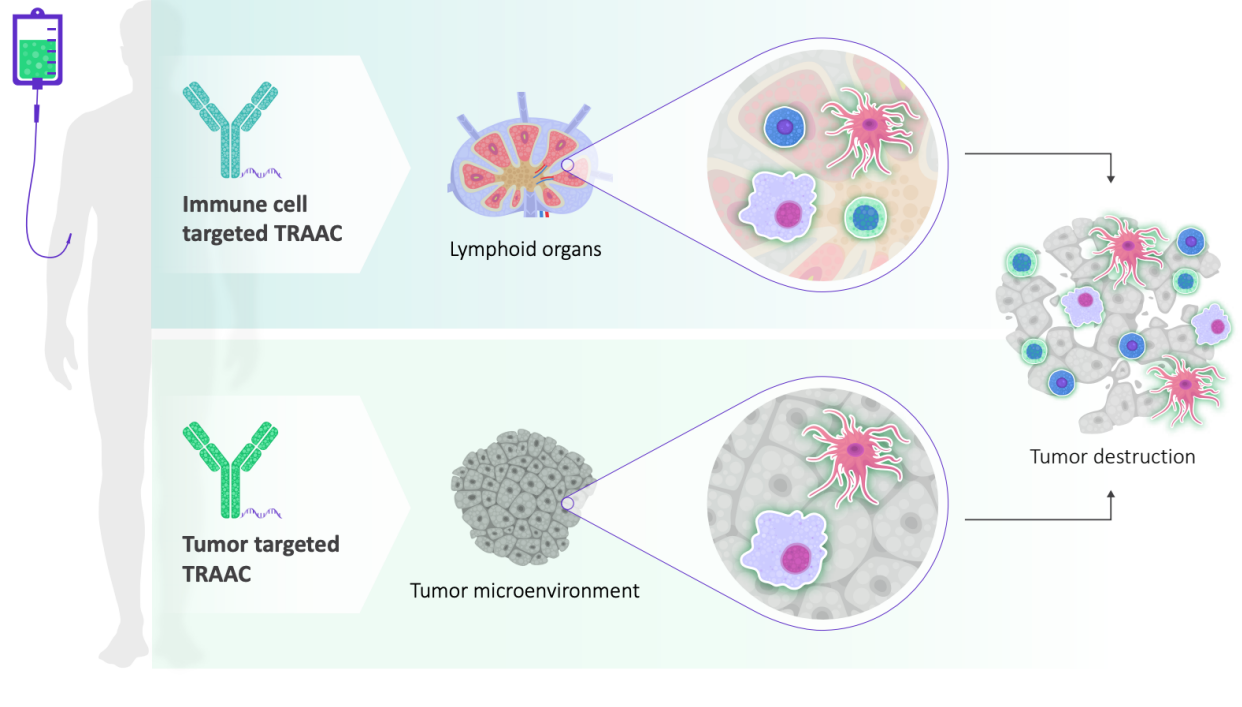
Immune cell targeted TRAAC
Lymphoid organs
Tumor destruction
Tumor targeted TRAAC
Tumor microenvironment

References
1. Kobold., Innate and adaptive immunity combined for cancer treatment. *PNAS* 2019, 116 (4) 1087-1088.
2. Dowling et al., Toll-like receptors: the swiss army knife of immunity and vaccine development. *Clin. Transl. Immunology*. 2016 *5*(5), e85–10.
3. Hamid et al., Intratumoral Immunotherapy—Update 2019. *The Oncol*. 2019; 25:343–359.

Immune cell targeting TRAAC

Fig 2: T-CpG is a potent TLR9 agonist with targeted activation across species

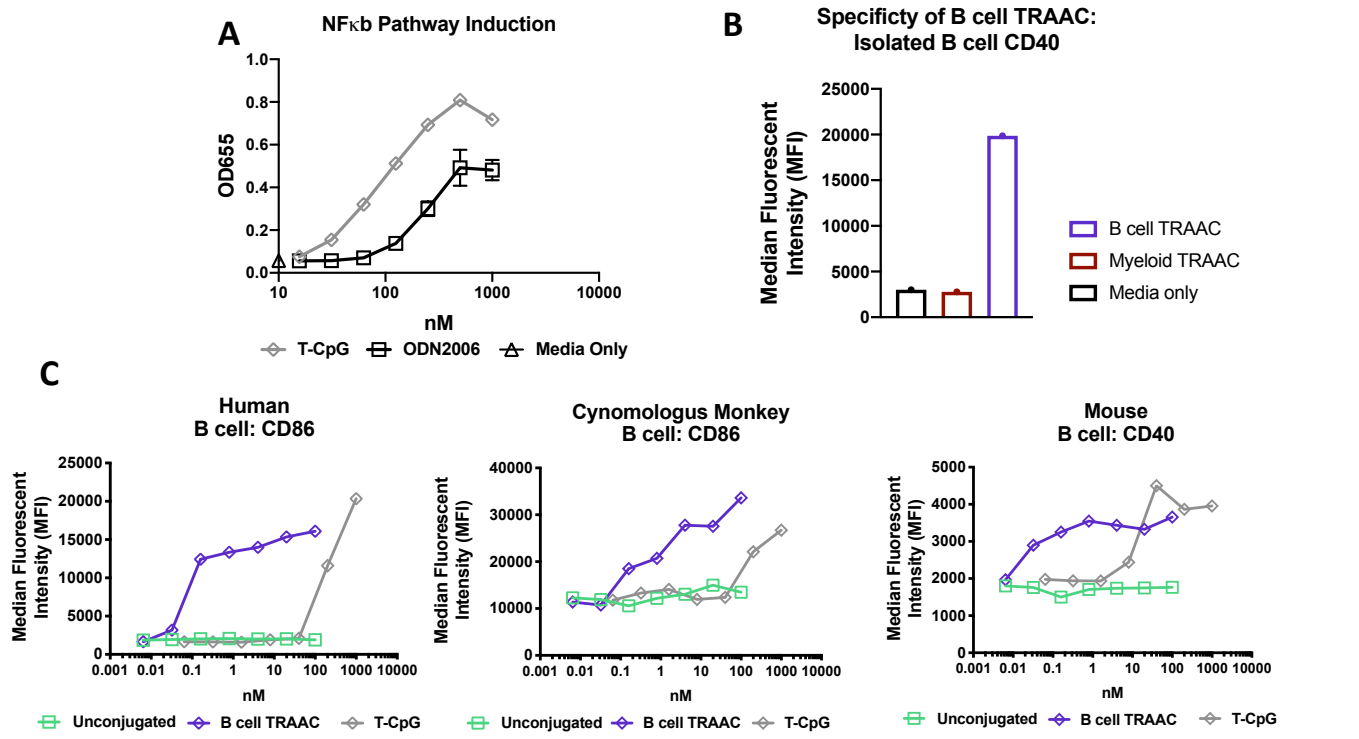
A
NFκb Pathway Induction
OD655
nM
T-CpG
ODN2006
Media Only
B
Specificty of B cell TRAAC: Isolated B cell CD40
Median Fluorescent Intensity (MFI)
B cell TRAAC
Myeloid TRAAC
Media only
C
Human B cell: CD86
Cynomologus Monkey B cell: CD86
Mouse B cell: CD40
Unconjugated
B cell TRAAC
T-CpG

Figure 2: NFκB reporter cells were stimulated with T-CpG or ODN2006 for 18hrs (A). Isolated human B cells were stimulated for 48hrs with B cell or myeloid TRAAC, surface marker expression was assayed by flow cytometry (B). Human, cynomolgus PBMCs, and mouse splenocytes were stimulated with B cell TRAAC, or T-CpG for 48hrs and expression of activating receptors were assayed by flow cytometry (C).

Fig 3: TRAACs directed to human pDC, myeloid and B cells induce potent targeted cellular activation and cytokine production

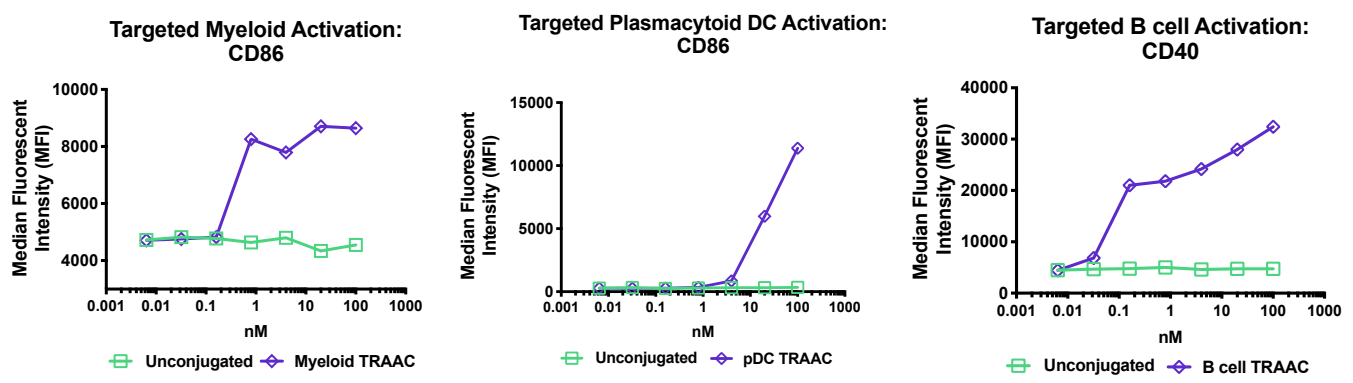
Targeted Myeloid Activation: CD86
Targeted Plasmacytoid DC Activation: CD86
Targeted B cell Activation: CD40
Median Fluorescent Intensity (MFI)
nM
Unconjugated
Myeloid TRAAC
pDC TRAAC
B cell TRAAC

Cell type	TLR9 activation
pDC	✓
B cell	✓
Myeloid	✓

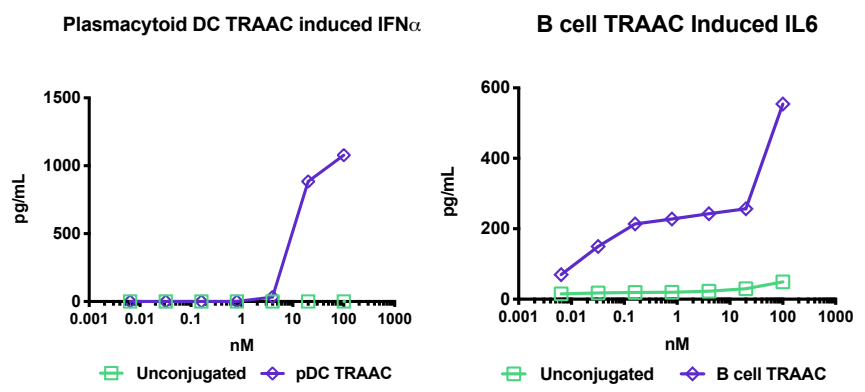
Plasmacytoid DC TRAAC induced IFNα
B cell TRAAC Induced IL6
pg/mL
nM
Unconjugated
pDC TRAAC
B cell TRAAC

Figure 3. Human PBMCs were stimulated for 48hrs in the presence of pDC, myeloid and B cell-directed TRAACs. Surface marker expression and cytokines was assayed by flow cytometry or ELISA.

Fig 4: TRAACs elicit indirect activation of non-targeted immune cells

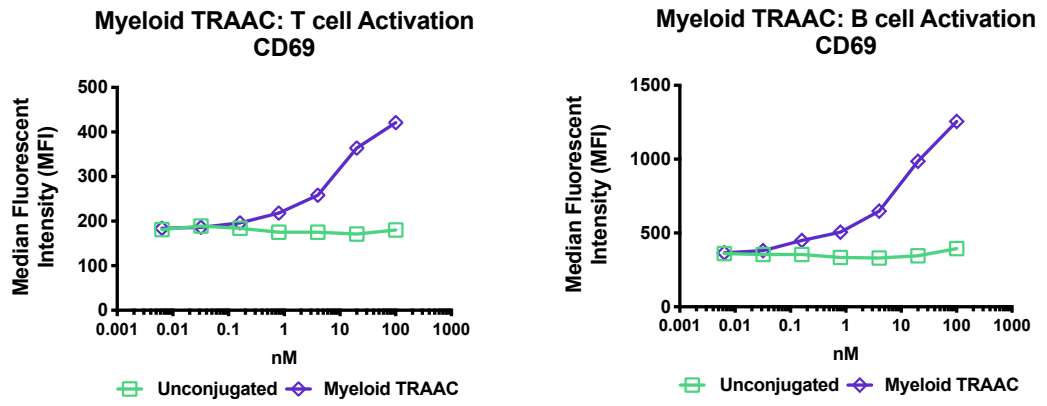
Myeloid TRAAC: T cell Activation CD69
Myeloid TRAAC: B cell Activation CD69
Median Fluorescent Intensity (MFI)
nM
Unconjugated
Myeloid TRAAC

Figure 4. Human PBMCs were stimulated with myeloid TRAAC for 48hrs and assayed for cell markers of activation of non-targeted T cells and B cells by flow cytometry.

Fig 5: Systemic administration of immune cell targeted TRAACs exhibit potent, dose-dependent, single agent activity in syngeneic tumor models

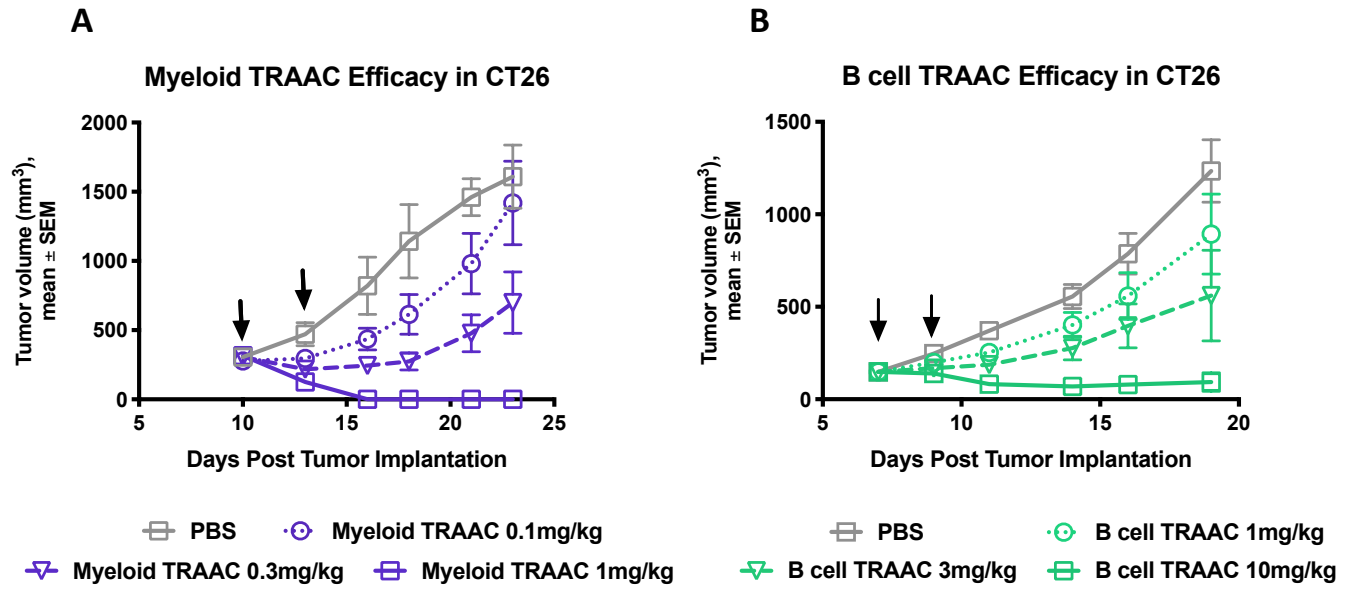
A
Myeloid TRAAC Efficacy in CT26
B
B cell TRAAC Efficacy in CT26
Tumor volume (mm³), mean ± SEM
Days Post Tumor Implantation
PBS
Myeloid TRAAC 0.1mg/kg
Myeloid TRAAC 0.3mg/kg
Myeloid TRAAC 1mg/kg
B cell TRAAC 1mg/kg
B cell TRAAC 3mg/kg
B cell TRAAC 10mg/kg

Figure 5. Mice bearing CT26 were intraperitoneally (i.p.) treated with PBS or myeloid TRAAC at 0.1mg/kg, 1mg/kg or 3mg/kg (A) or B cell TRAAC at 1mg/kg, 3mg/kg or 10mg/kg (B). Murine TRAACs are conjugated to mouse reactive T-CpG. Arrows indicate doses administrated.

Fig 6: Durable anti-tumor memory response in tumor free syngeneic model following administration of immune cell targeted TRAAC

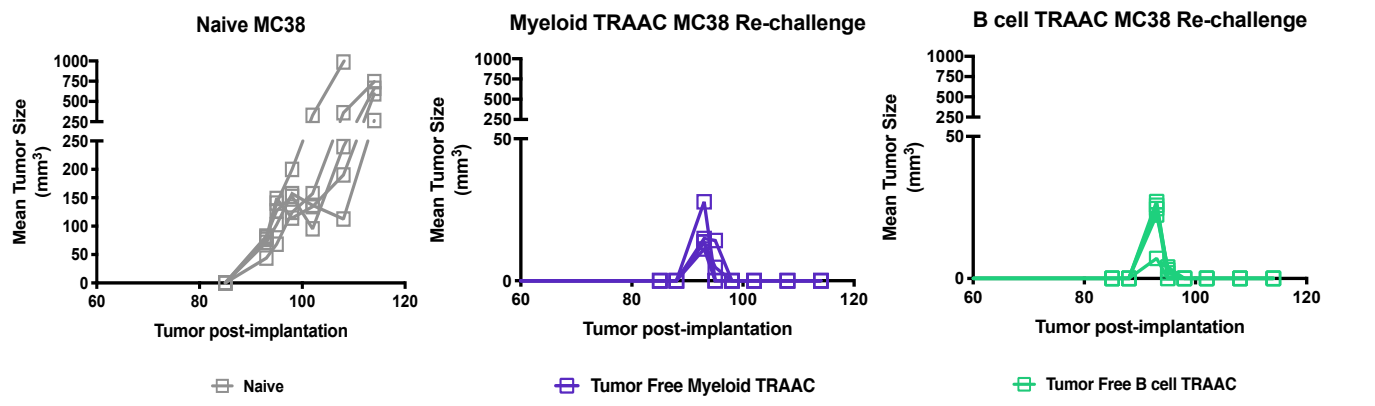
Naive MC38
Myeloid TRAAC MC38 Re-challenge
B cell TRAAC MC38 Re-challenge
Mean Tumor Size (mm³)
Tumor post-implantation
Naive
Tumor Free Myeloid TRAAC
Tumor Free B cell TRAAC

Figure 6. Tumor eradicated mice from MC38 bearing mice following 10mg/kg 2q3 systemic treatment with either myeloid TRAAC or B cell TRAAC were re-challenged 60-70 days post-initial tumor clearance. Naïve age-matched mice were used as control for tumor growth.

Fig 7: TRAAC evokes rapid innate and adaptive immune signatures within tumor microenvironment

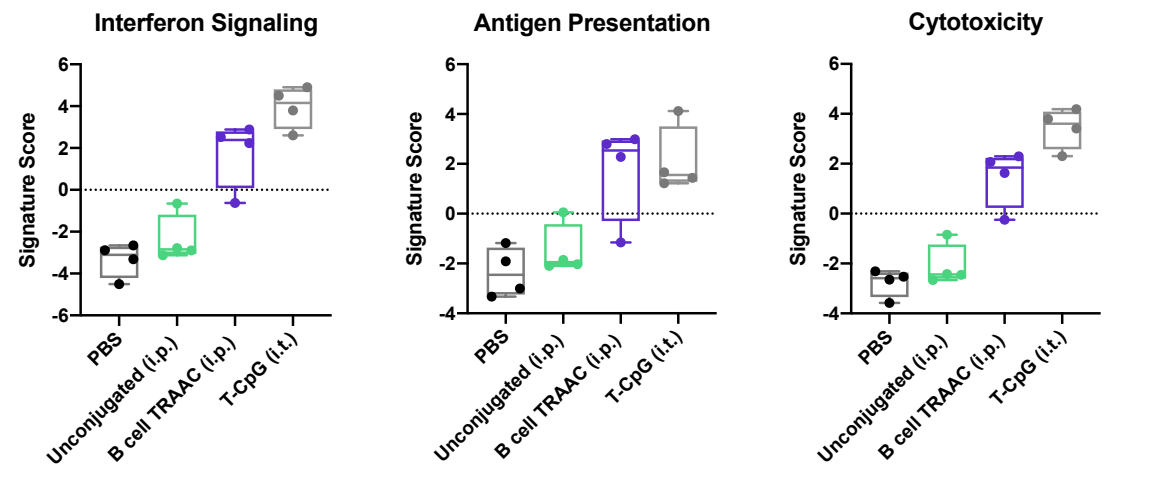
Interferon Signaling
Antigen Presentation
Cytotoxicity
Signature Score
PBS
Unconjugated (i.p.)
B cell TRAAC (i.p.)
T-CpG (i.t.)

Figure 7. Mice bearing CT26 were dosed intraperitoneally (i.p.) once with B cell TRAAC or unconjugated antibody at 10mg/kg. Mouse reactive T-CpG was injected directly into the tumor at 50ug/mouse. Tumors were harvested 8hrs post-dose for nanostring analysis. Raw data was normalized and visualized using nsolver software by Nanostring.

Tumor Targeting TRAAC

Fig 8: HER2-directed TRAAC induces tumor antigen dependent myeloid activation

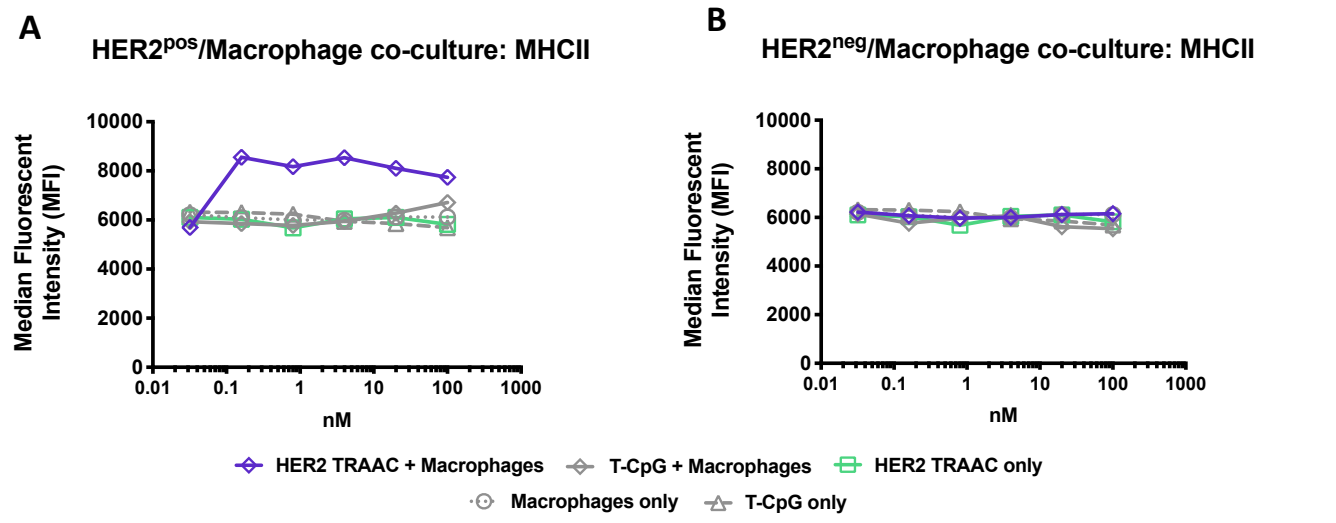
A
HER2pos/Macrophage co-culture: MHCII
B
HER2neg/Macrophage co-culture: MHCII
Median Fluorescent Intensity (MFI)
nM
HER2 TRAAC + Macrophages
T-CpG + Macrophages
HER2 TRAAC only
Macrophages only
T-CpG only

Figure 8. Bone marrow-derived mouse macrophages were co-cultured in the presence of HER2^{pos} (A) or HER2^{neg} tumors (B) and treated with HER2 TRAAC or mouse reactive T-CpG for 24hrs. Median MHCII was assessed by flow cytometry.

Fig 9: HER2-directed TRAAC demonstrates potent and durable anti-tumor response in syngenetic tumor model

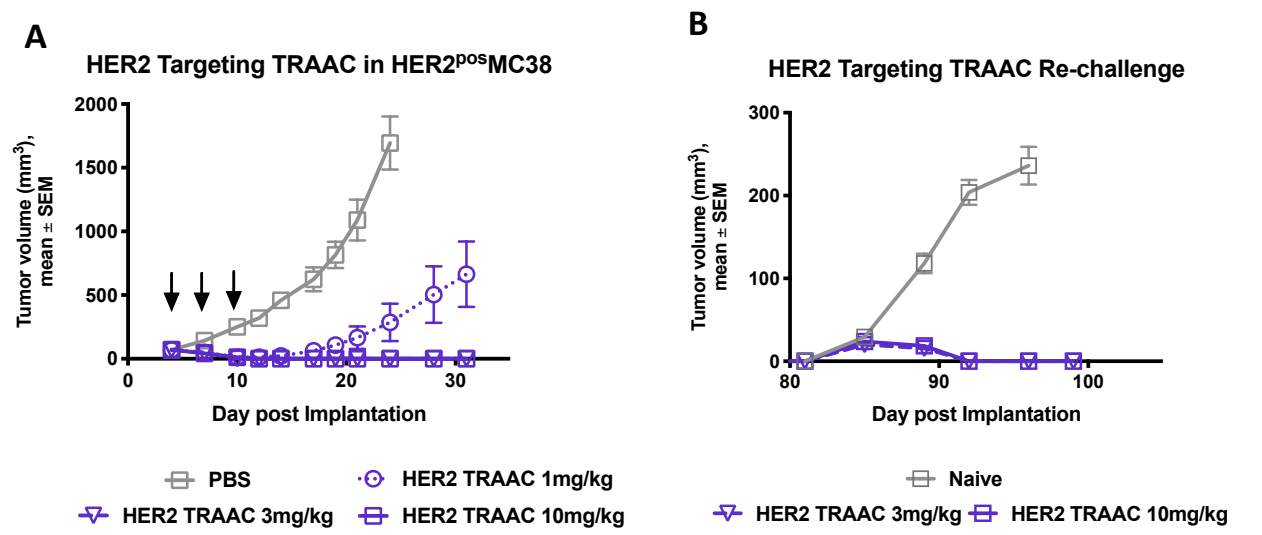
A
HER2 Targeting TRAAC in HER2posMC38
B
HER2 Targeting TRAAC Re-challenge
Tumor volume (mm³), mean ± SEM
Day post Implantation
PBS
HER2 TRAAC 1mg/kg
HER2 TRAAC 3mg/kg
HER2 TRAAC 10mg/kg
Naive

Figure 9. Mice bearing HER2^{pos} MC38 were intraperitoneally (i.p.) treated with HER2-directed TRAAC at 1mg/kg, 3mg/kg or 10mg/kg (A). Arrows indicate doses administrated. Tumor eradicated mice were rechallenged with HER2^{pos}MC38 60 days post-initial tumor clearance, naïve mice were used as control for tumor growth (B).

Exploratory Studies in Cynomolgus Monkey

- Immune targeting TRAACs were well tolerated following repeated intravenous injections in cynomolgus monkeys, as assessed by clinical observations, body weights, food consumption, hematology, serum chemistry and histopathology.

Conclusions

- TRAAC is a novel immune activating platform that engages innate and adaptive immunity.
- TRAAC delivers T-CpG, a potent differentiated TLR9 agonist, to targeted immune cells or tumor microenvironment leading to immune activation.
- TRAACs demonstrate robust and durable tumor regression in multiple syngeneic tumor models, as a single agent.
- TRAACs exhibit favorable tolerability and safety profiles in cynomolgus monkeys.

- **Designed as a systemic therapy, TRAACs demonstrate potent targeted immune activation, robust anti-tumor activity and favorable tolerability in pre-clinical settings.**
- **TRAAC platform has broad therapeutic potentials against multiple solid tumor malignancies.**